Antibody-armed oncolytic Vaccinia virus to block immunosuppressive pathways in the tumor microenvironment

Jean-Baptiste Marchand[1], Monika Semmrich[2], Laetitia Fend[1], Ulla-Carin Tornberg[2], Nathalie Silvestre[1], Björn Frendéus[2], Eric Quéméneur[1]
[1] Transgene S.A., Illkirch-Graffenstaden, France
[2] BioInvent International A.B., Lund, Sweden

ABSTRACT

***Vaccinia* virus (VV)** has proven to be a powerful oncolytic vector thanks to its large spectrum of tumor cell targets, large genome capacity, good safety properties, and strong immunogenic properties. We have developed an improved VV-based platform in the Copenhagen strain, with a **double deletion in the J2R/TK, and I4L/RR genes**, that displays a very high therapeutic index (over 10^4), compatible with the intravenous route.
VV_{TK-RR-} can be genetically modified to **express full-length monoclonal** antibodies, and additional cytokines, at the site of active viral replication and accumulation, i.e. directly in the tumor. Thus, checkpoint blockers can be targeted to boost the immune response locally initiated by the oncolytic activity. Moreover, the benefit of vectorizing a mAb would be to **diminish the severe side effects** reported for some systemic administration, and to **increase the intratumoral antibody concentration** to maximize the probability of efficacy.
Our first products addressed the two major inhibitory pathways : a VV-anti-PD-1 to restore the activity of infiltrated effector T cells, and a **VV-anti-CTLA-4 to selectively deplete intratumoral regulatory T cells via ADCC.**

ACHIEVEMENTS

- **Combination of ICI and oncolytic VV treatments are additive/synergic**
- **Functional ICI (full IgG against mPD-1, i.e. mAb and simpler formats) can be vectorized in oncolytic vaccinia virus.**
- **Therapeutic molecule accumulate into the tumor while the circulating concentrations are low compared to a regular combination**
- **Therapeutic efficacy of a vectorized ICI comparable to that obtained with a regular combination with low systemic exposure .**

INVIRIO PLATFORM MAIN FEATURES

Main features of Transgene's vaccinia oncolytic virus platform:

- Good safety profile and high therapeutic index
- Thanks to vaccination experience, strong track record of clinical use
- Pure cytoplasmic replication (no risk for genome integration or mutagenesis)
- Good immunological balance (Th1 vs Th2, anti-tumor vs anti-viral responses, etc.)
- Well-established processes for GMP manufacturing
- Large DNA insertions are possible (up to 25 kb) as several different expression cassettes
 - enzymes, cytokines, antibodies … have been successfully vectorized
- Thymidine kinase (TK) and Ribonucleotide Reductase (RR) double deleted
 - restrict replication of vaccinia virus to proliferative cells (*e.g.* tumoral cells): safer than WT vaccinia virus
- Copenhagen strain: best oncolytic activity among vaccinia virus
 - used as surrogate oncolytic vaccinia virus for *in vivo* preclinical studies

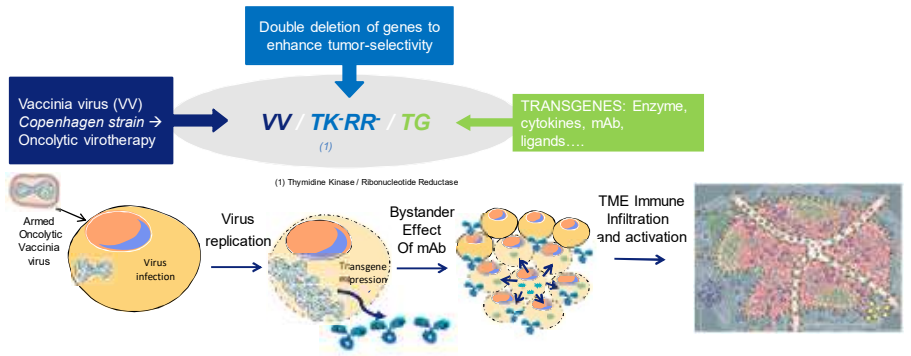
Double deletion of genes to enhance tumor-selectivity
Vaccinia virus (VV)
Copenhagen strain →
Oncolytic virotherapy
VV / TK-RR- / TG
(1)
TRANSGENES: Enzyme, cytokines, mAb, ligands….
(1) Thymidine Kinase / Ribonucleotide Reductase
Armed Oncolytic Vaccinia virus
Virus infection
Virus replication
Transgene expression
Bystander Effect Of mAb
TME Immune Infiltration and activation

VV-@PD-1 AS A CASE STUDY

- Anti-murine PD-1 (J43) has been vectorized as mAb, Fab or scFv
- two promoters were used to express heavy and light chains (pH5R is a stronger promoter than p7.5K) of Fab and mAb
- Fabs and scFv have been His-tagged
- Here Western reserve (WR) vaccinia strain used because of its better replication in murine cells

vaccinia viruses expressing anti-mPD-1 blockers

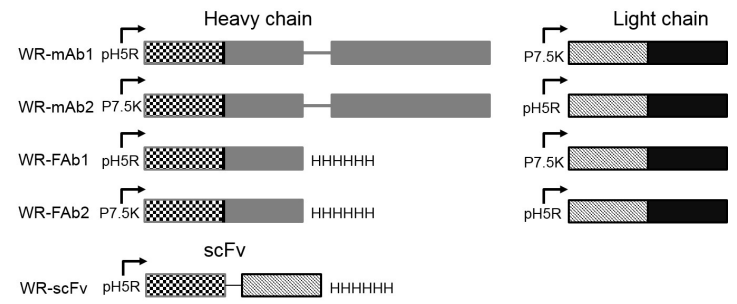
Heavy chain
Light chain
WR-mAb1 pH5R
P7.5K
WR-mAb2 P7.5K
pH5R
WR-FAb1 pH5R HHHHHH
P7.5K
WR-FAb2 P7.5K HHHHHH
pH5R
scFv
WR-scFv pH5R HHHHHH

References

- **Foloppe *et al.* Gene Ther. 2008 Oct;15(20):1361-71**
- **Fend *et al* Cancer Res. 2017 Aug 1;77(15):4146-4157**
- **Kleinpeter *et al.* Oncoimmunology. 2016 Sep 9;5(10):e1220467**
- **WO2009/065546 Poxviral Oncolytic Vectors**
- **WO2016/009017 Combination of oncolytic virus with immune checkpoint modulators**
- **WO2016/008976 Oncolytic virus for expression of immune checkpoint modulators**

RESULTS

1: ICI combination with oncolytic VV improved the anti-tumoral efficacy

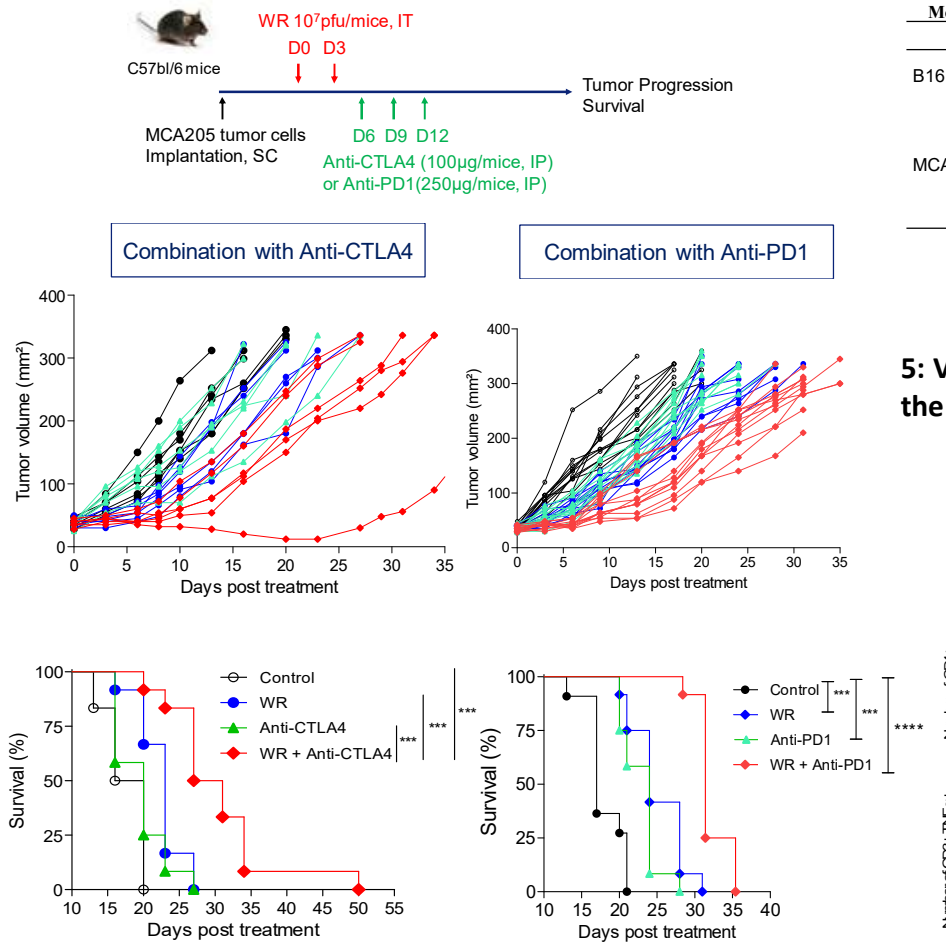
C57bl/6 mice
WR 10⁷pfu/mice, IT
D0 D3
Tumor Progression
Survival
MCA205 tumor cells Implantation, SC
D6 D9 D12
Anti-CTLA4 (100µg/mice, IP)
or Anti-PD1(250µg/mice, IP)
Combination with Anti-CTLA4
Combination with Anti-PD1
Tumor volume (mm²)
Days post treatment
Survival (%)
Control
WR
Anti-CTLA4
WR + Anti-CTLA4
Anti-PD1
WR + Anti-PD1

2: The purified vectorized mAb1, Fab1, scFv are functional

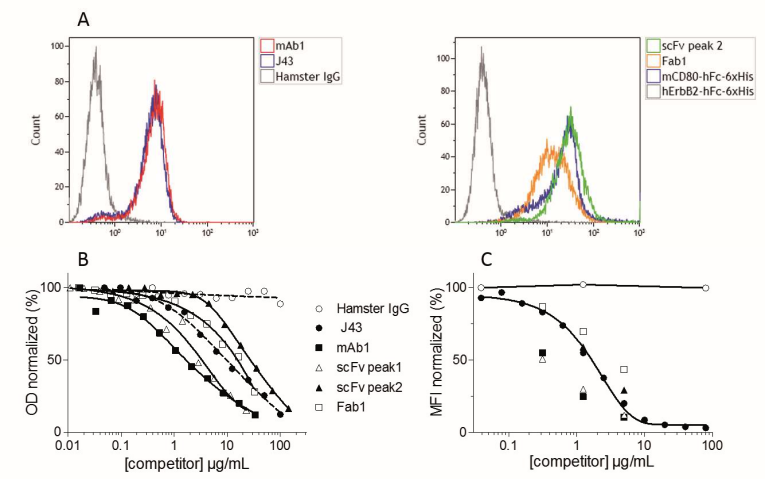
A
mAb1
J43
Hamster IgG
Count
scFv peak 2
Fab1
mCD80-hFc-6xHis
hErbB2-hFc-6xHis
B
C
OD normalized (%)
MFI normalized (%)
Hamster IgG
J43
mAb1
scFv peak1
scFv peak2
Fab1
[competitor] µg/mL

Binding of purified mAb1, Fab1 and scFv to mPD-1-positive EL4 cells (A). Blocking of mPD1 –mPD-L1 interaction by purified mAb1, Fab1 and scFv monitored by either ELISA (B) or flow cytometry (C) assays.

3: Vectorization allows the accumulation of anti-mPD1 into the tumor with a low systemic exposition

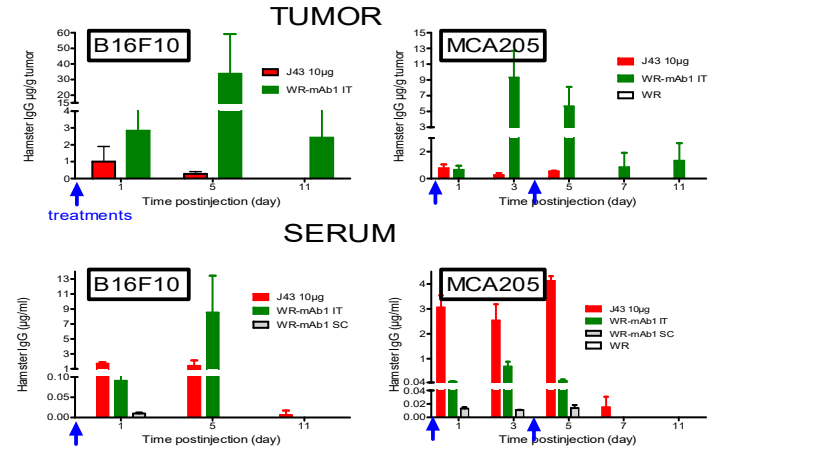
TUMOR
B16F10
MCA205
Hamster IgG µg/g tumor
J43 10µg
WR-mAb1 IT
WR
Time postinjection (day)
treatments
SERUM
Hamster IgG (µg/ml)
WR-mAb1 SC

Concentrations of anti-mPD1 measured in tumor homogenates (top) or in sera (bottom) in two tumor models B16F10 (left) and MCA205 (right) after I.T injections (bleu arrows) of either WR-mAb1 (10^7 pfu) or 10 µg of J43. WR-mAb1 was also injected in absence of tumor (SC) to illustrate the low replication in non-tumoral tissue.

4: Tumor/Serum ratio of mAb demonstrate the tumor accumulation only in case of vectorization

Models	treatments	Day post-injection 1	3	5	7	11
B16F10	J43	0.6	-	0.2	-	3*
	WR-mAb1	31	-	4	-	605*
MCA 205	J43	0.2	0.1	0.1	4*	ND
	WR-mAb1	8	13	51	84*	132*

If one of the concentrations was below the LOQ, LOQ was used to calculate the ratio which appears labelled with *. (NA: not applicable, ND: not detected: both concentrations in tumor and serum were below LOQ)..

5: Vaccinia virus induces a massive immune infiltration in to the tumor

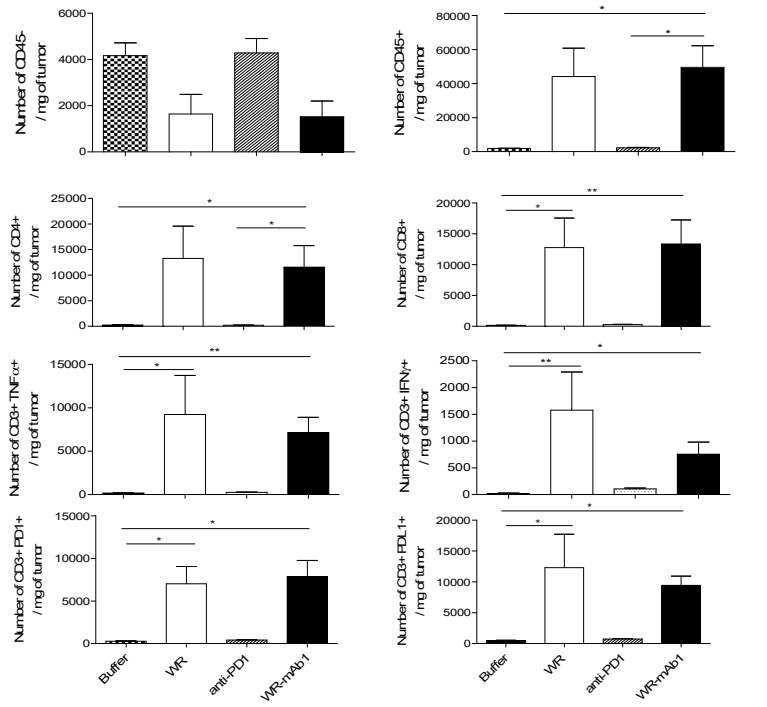
Number of CD45- / mg of tumor
Number of CD45+ / mg of tumor
Number of CD4+ / mg of tumor
Number of CD8+ / mg of tumor
Number of CD3+ TNFα+ / mg of tumor
Number of CD3+ IFNγ+ / mg of tumor
Number of CD3+ PD1+ / mg of tumor
Number of CD3+ PDL1+ / mg of tumor
Buffer
WR
anti-PD1
WR-mAb1

Analysis of the MCA205 tumor immune infiltrate 3 days after last treatment in the conditions presented in chapter 3 . No difference was observed between the immune profile induced by WR and WR-mAb1 treatments.

6: Two vectorized formats of anti-PD1 inhibit tumor growth with equivalent efficacy to combination of WR "empty" + 3x250 µg of anti-mPD1

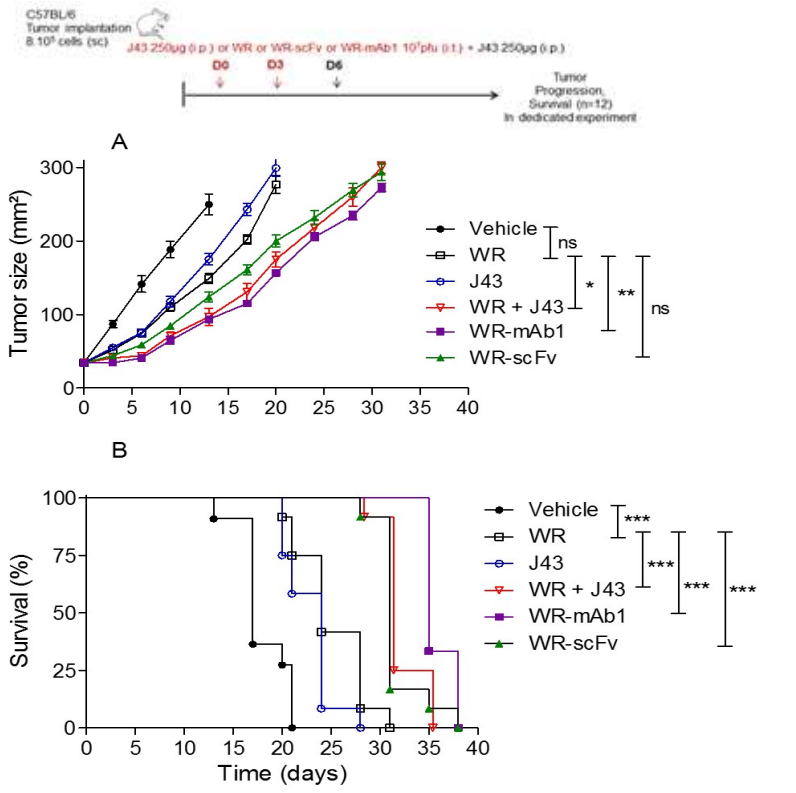
D0
D3
D6
Tumor Progression, Survival (n=12) In dedicated experiment
A
Tumor size (mm²)
Vehicle
WR
J43
WR + J43
WR-mAb1
WR-scFv
ns
B
Survival (%)
Time (days)

Antitumoral efficacy in the MCA205 model of WR-mAb1 and WR-scFv after 2 injections IT. WR-mAb1 and WR-scFv efficacy was benchmarked to the WR (without transgene) or to anti-PD1 alone (3 injections IP of 250µg) or to the combination of WR + J43. Results are represented as the mean tumor size (A) or as survival percentage (B).

www.transgene.fr SITC congress, WASHINGTON, D.C 2018 Transgene, 400 Boulevard Gonthier d'Andernach - Parc d'Innovation - CS80166 67405 Illkirch Graffenstaden Cedex - France @TransgeneSA